ANV419 is a novel CD122-selective IL-2/anti-IL-2 antibody fusion protein with potent CD8+ T cell and NK cell stimulatory function *in vitro* and *in vivo*

Christoph Huber[1], Barbara Branetti[2], Jean-Michel Rondeau[2], Simone Popp[2], Catherine Regnier[2], Daniel Kaiser[2], Patrizia Murer[1], Laetitia Petersen[1] and Andreas Katopodis[1]
[1]Anaveon, Basel Switzerland, [2]Novartis Institutes for Biomedical Research, Basel Switzerland

Background

ANV419 is a uniquely engineered IL-2 fusion to an antibody which promotes signaling through the dimeric IL-2 receptor (IL-2Rβ/γ, CD122/CD132) but not through the trimeric receptor (IL-2Rα/β/γ, CD25/CD122/CD132). It preferentially stimulates the proliferation of CD8+ T cells and NK cells while avoiding the proliferation of immunosuppressive regulatory T cells (Treg). Therefore, ANV419 has the potential to substantially separate CD8+ T-cell and NK cell proliferation and anti-tumor responses from the dose limiting toxicities of recombinant IL-2 (aldesleukin). ANV419 has antibody like stability and behavior and is currently in late preclinical development for tumor immunotherapy.

Results

ANV419 is a fusion protein of IL-2 and an IL-2 specific antibody that binds with high affinity to the IL-2Rα binding domain of IL-2. IL-2 is fused to the light chain CDR domain of the antibody, allowing ANV419 to present IL-2 to the dimeric receptor while sterically excluding IL-2Rα as shown in the crystal structure.

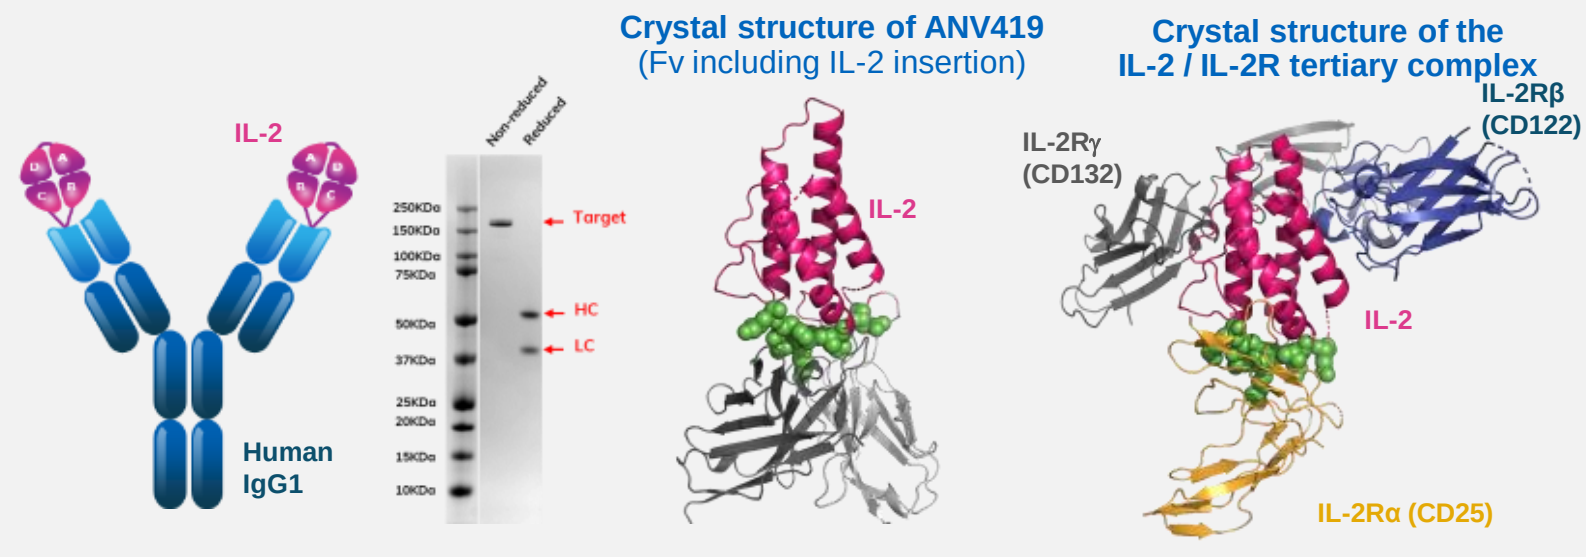
IL-2
Human IgG1
Non-reduced
Reduced
250KDa
150KDa
100KDa
75KDa
50KDa
37KDa
25KDa
20KDa
15KDa
10KDa
Target
HC
LC
Crystal structure of ANV419
(Fv including IL-2 insertion)
IL-2
Crystal structure of the IL-2 / IL-2R tertiary complex
IL-2Rβ (CD122)
IL-2Rγ (CD132)
IL-2
IL-2Rα (CD25)

ANV419 is an antibody-like protein **IL-2 residues involved in CD25 binding are buried in ANV419 (shown in green)**

ANV419 does not bind to IL-2Rα and retains binding to IL-2Rβ with affinity comparable to IL-2

Binding of ANV419 to the IL-2Rα and IL-2Rβ chains was measured by Biacore™ surface plasmon resonance. ANV419 binds to the IL-2Rβ chain with an affinity equal or better than recombinant human IL-2 or IL-2-Fc fusion protein. As expected from the crystal structure, ANV419 does not bind to the IL-2Rα chain.

	K_D IL-2Rα (CD25)	K_D IL-2Rβ (CD122)
ANV419	No binding	377 nM
IL-2-Fc	11.7 nM	1490 nM
IL-2 (published)	10 nM	~500 nM

ANV419 has reduced potency for STAT5 phosphorylation in Treg cells while maintaining full potency in CD8 T cells

Human PBMCs were incubated with ANV419 or IL-2 (Proleukin ®) for 10 min and phosphorylation of STAT5 was measured in Tregs and CD8+ T cells by flow cytometry (n=3 donors). Compared to Proleukin ®, the potency for STAT5 phosphorylation of ANV419 is reduced greater than 1000-fold in Treg compared to Proleukin ®. ANV419 retains full potency comparable to Proleukin ® for activation of CD8+ T cells.

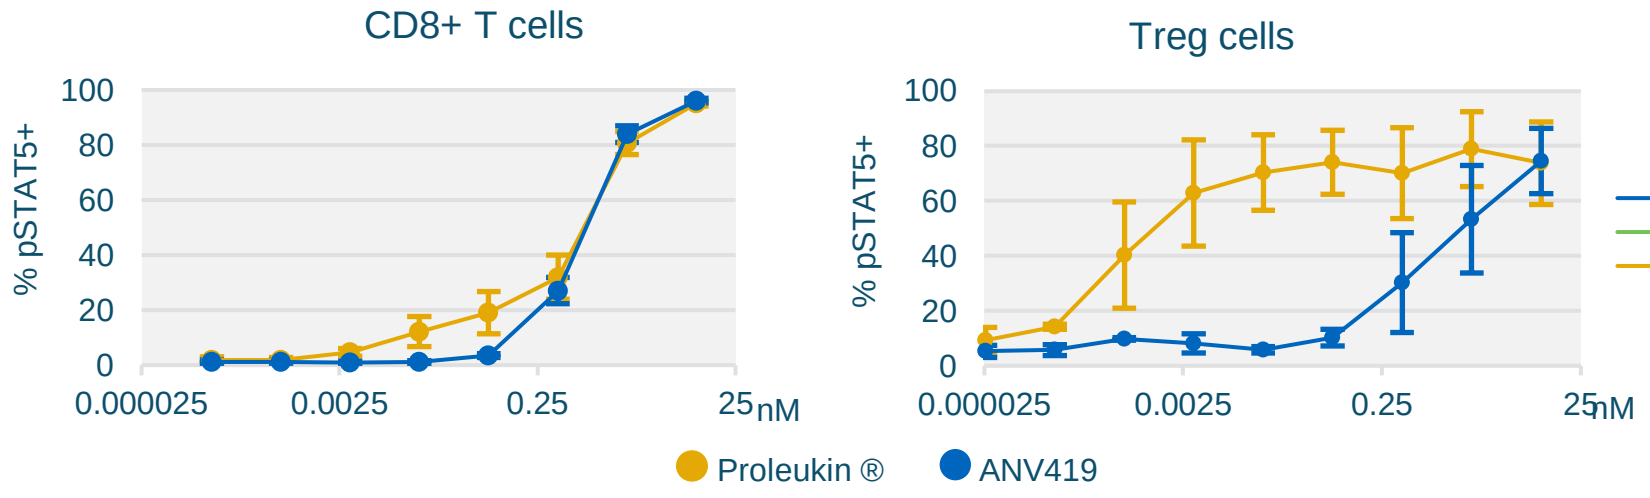
CD8+ T cells
Treg cells
% pSTAT5+
0.000025
0.0025
0.25
25
nM
Proleukin ®
ANV419

ANV419 selectively induces proliferation of CD8+ T cells and NK cells but not CTLL-2 cells that express the trimeric IL-2Rα/β/γ

Human CD8+ T cells and NK cells were purified from PBMCs, labeled with CFSC and cultured for 8 days. Proliferation was measured by determining the extent of CFSC dilution. Mouse CTLL2 cells were cultured for 48 hr at the indicated concentration of IL-2 or ANV419, proliferation was quantified by incubation with WST-1 for 4 hr and measurement of OD at 450 nm.

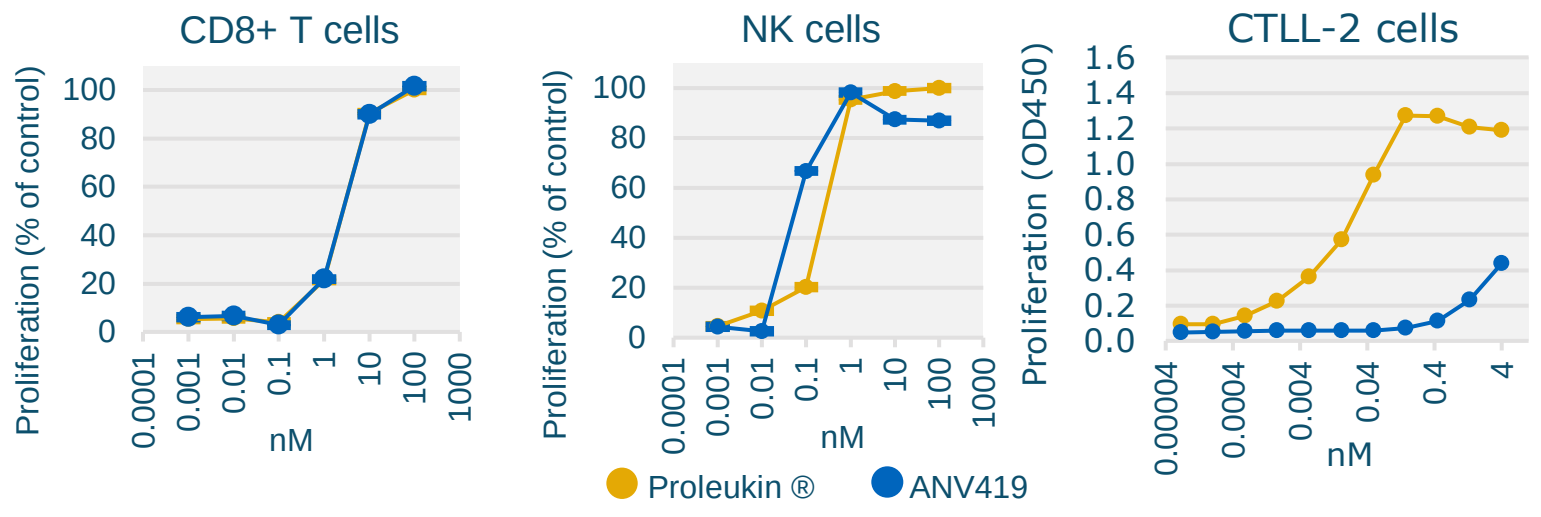
CD8+ T cells
NK cells
CTLL-2 cells
Proliferation (% of control)
Proliferation (OD450)
nM
Proleukin ®
ANV419

In mice ANV419 expands CD8+ T cells and NK cells in a dose dependent manner with selectivity against Tregs up to 220 µg/kg

Single doses of ANV419 at 110, 220, and 440 µg/kg were administered to C57BL/6 mice i.p. Selective expansion of CD8+ T cells and NK cells in spleen correlates with selective induction of the proliferation marker Ki-67 in the same cells.

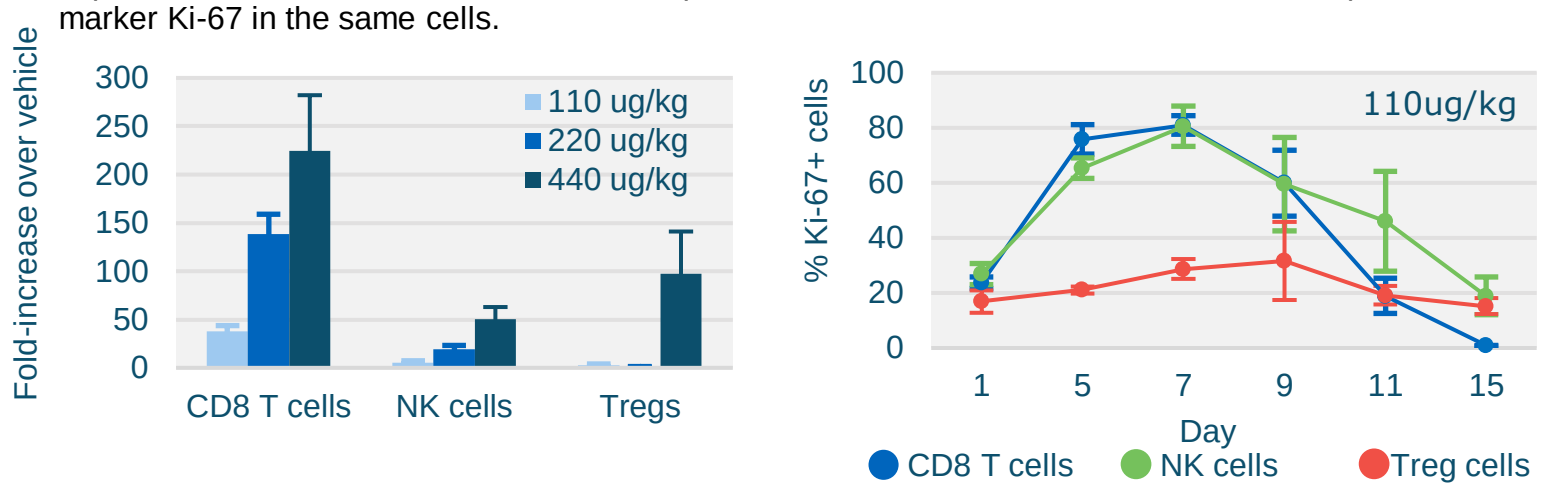
Fold-increase over vehicle
110 ug/kg
220 ug/kg
440 ug/kg
CD8 T cells
NK cells
Tregs
% Ki-67+ cells
110ug/kg
Day
CD8 T cells
NK cells
Treg cells

ANV419 inhibits tumor growth in CPI sensitive (H22) and resistant (B16F10, Renca) syngeneic mouse tumor models

Mice were inoculated with B16F10, Renca, or H22 syngeneic tumors. Treatment with ANV419 was initiated when the mean tumor size reached ~80-100mm². ANV419 was given i.v. at a dose of 200 µg/kg between 1 to 3x per week as indicated.

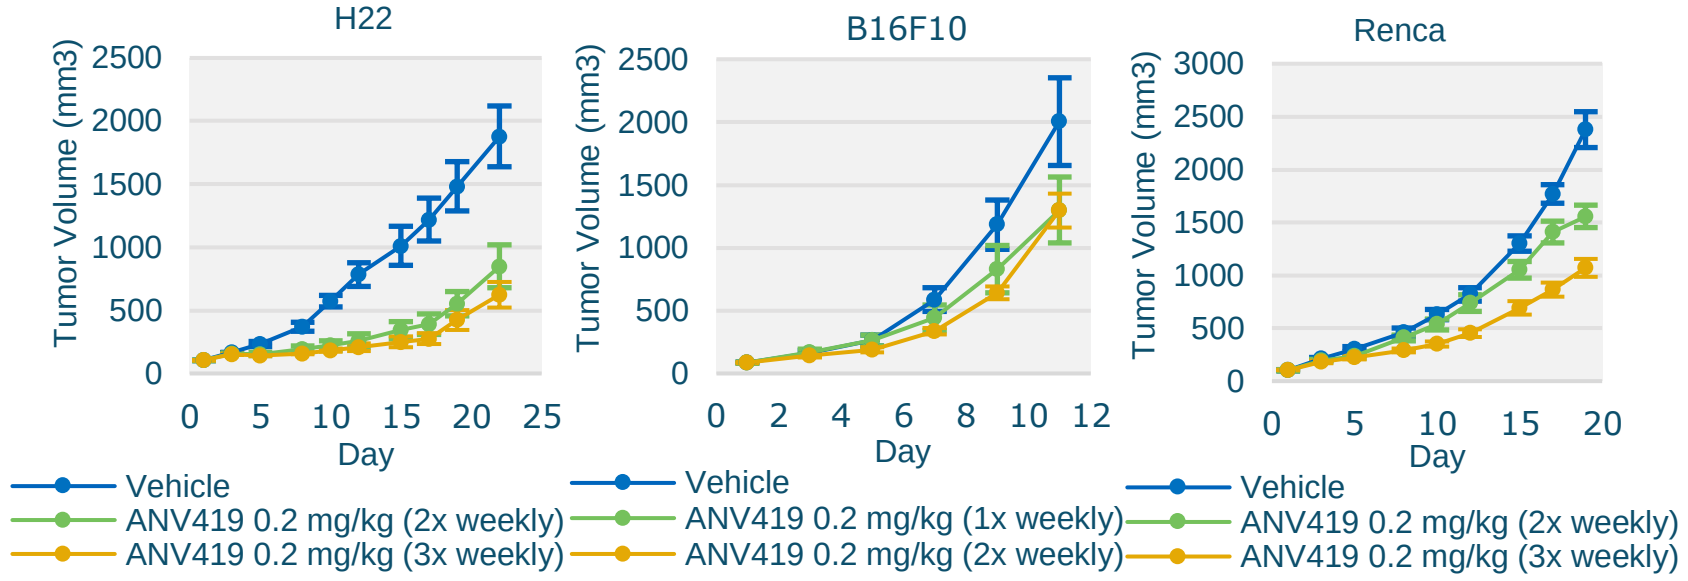
H22
B16F10
Renca
Tumor Volume (mm3)
Day
Vehicle
ANV419 0.2 mg/kg (2x weekly)
ANV419 0.2 mg/kg (3x weekly)
Vehicle
ANV419 0.2 mg/kg (1x weekly)
ANV419 0.2 mg/kg (2x weekly)
Vehicle
ANV419 0.2 mg/kg (2x weekly)
ANV419 0.2 mg/kg (3x weekly)

Tumor Model	Tumor Type	Mouse strain	Maximum TGI*	P-value
B16F10	Melanoma	C57BL/6	46%	0.144
Renca	Renal Cell Carcinoma	BALB/c	55%	<0.0001
H22	Hepatocellular Carcinoma	BALB/c	67%	<0.001

** TGI, tumor growth inhibition*

In non-human primates, dose-dependent response to ANV419 with preferential expansion of CD8+ T cells and NK cells

Cynomolgus monkeys (3 males and 3 females per group) were dosed with vehicle or 30µg/kg, 100µg/kg, or 300µg/kg ANV419 on day 1 and on day 15. Blood was taken at the indicated timepoints for immunophenotyping analysis of CD8+ T cells, NK cells, and Tregs. A similar dose-dependent increase in the absolute cell counts and proliferation of CD8+ T cells and NK cells was observed after both doses. Selective proliferation and expansion of CD8+ T cells and NK cells was observed at doses up to 100µg/kg. Sensitivity to ANV419 follows the sequence NK cells > CD8+ T cells > Treg.

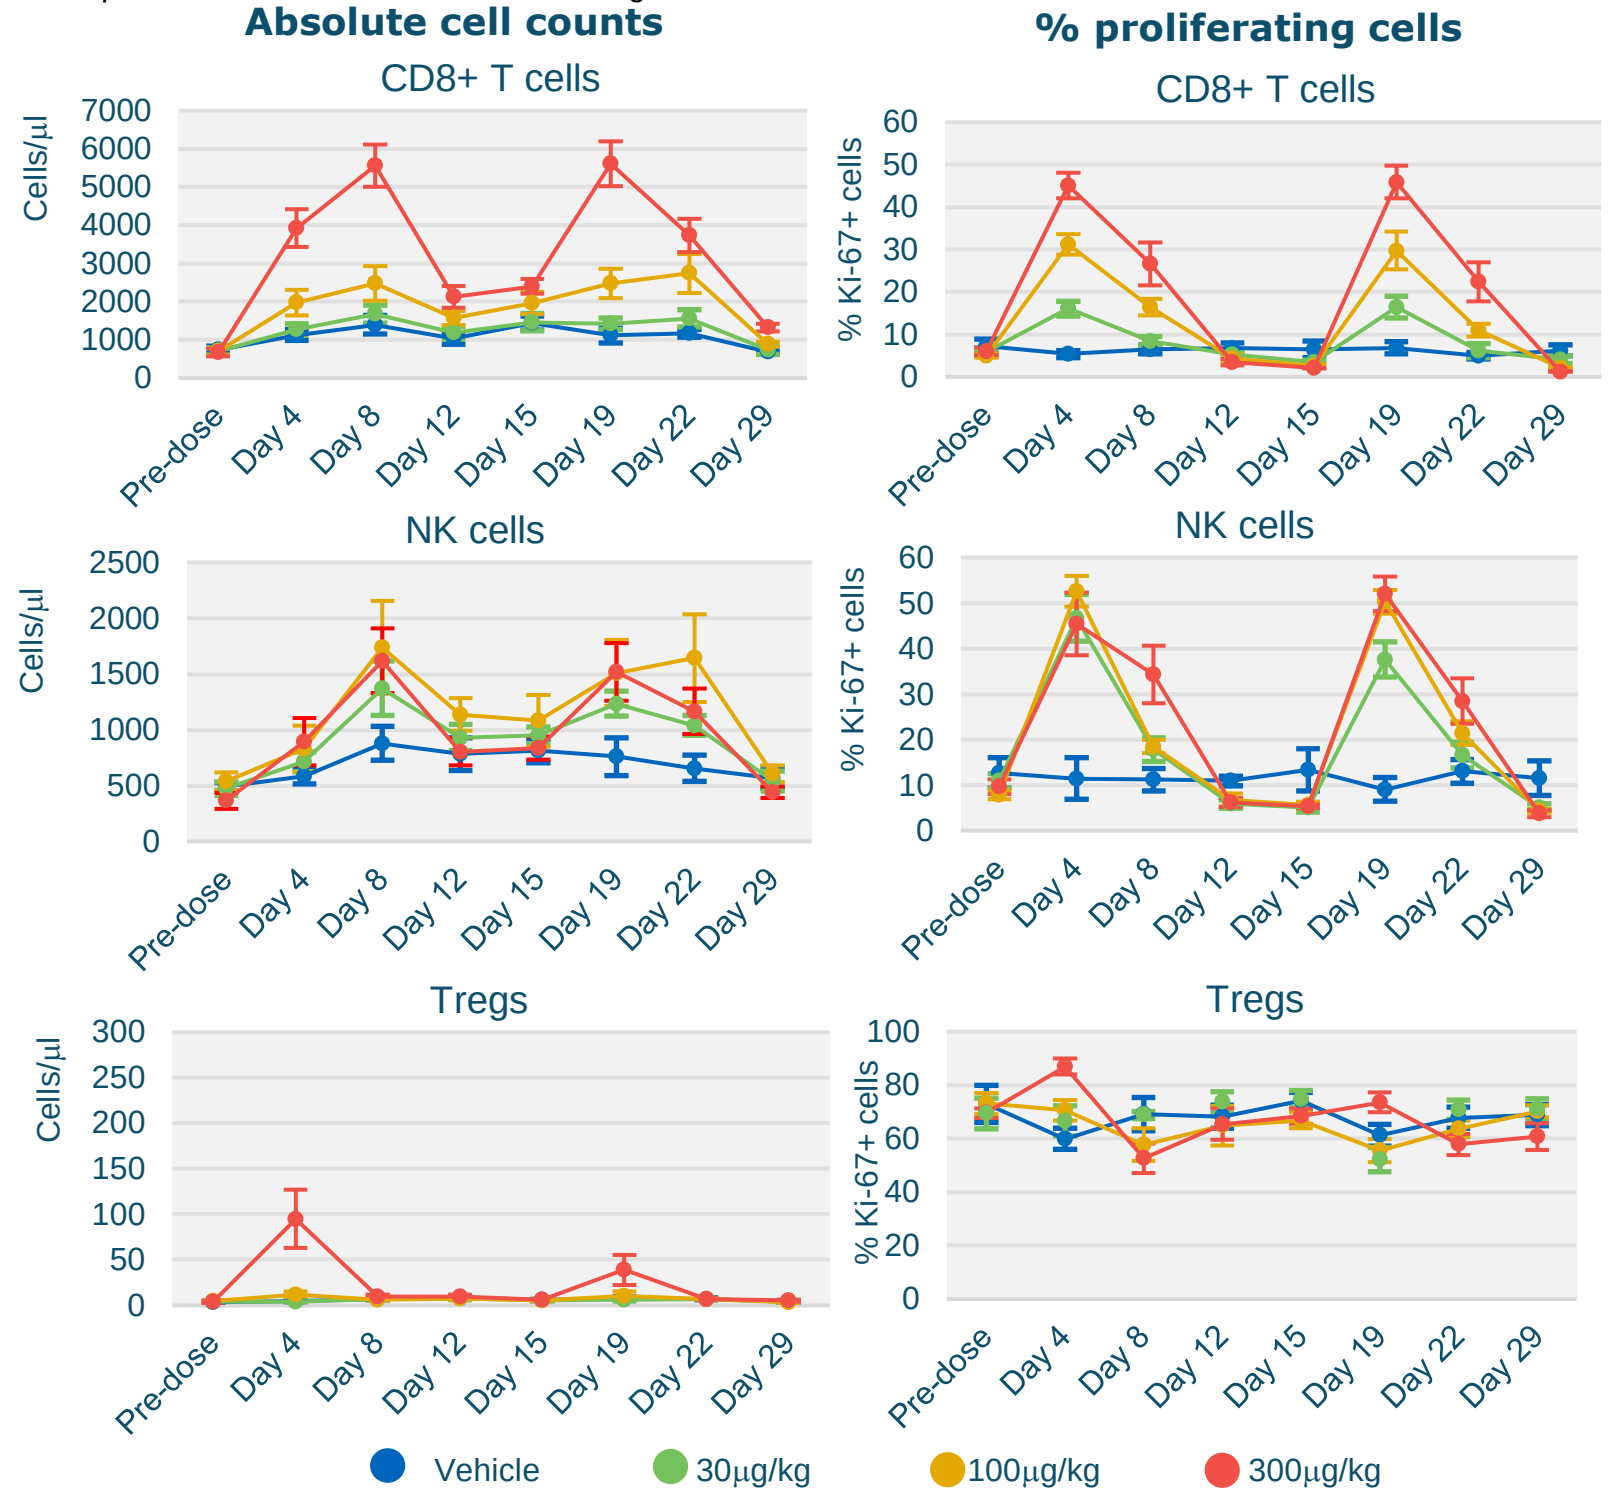
Absolute cell counts
% proliferating cells
CD8+ T cells
NK cells
Tregs
Cells/µl
% Ki-67+ cells
Pre-dose
Day 4
Day 8
Day 12
Day 15
Day 19
Day 22
Day 29
Vehicle
30µg/kg
100µg/kg
300µg/kg

ANV419 has a favorable safety profile in non-human primates

Expected IL-2 related pharmacology with increases of CD8+ T cells and NK cells without clinical signs of toxicity was observed at all doses tested. No infusion related adverse events or tolerability issues with short i.v. infusion over 1min were observed. No signs of vascular leak syndrome (body weight or clinical signs) or signs of cytokine release syndrome with no increase in IFNγ, TNFα, IL-6, and IL-8 over baseline were observed throughout the study.

Conclusions

ANV419 is a novel, CD122-biased, IL-2 immunocytokine with a unique structure and is both selective and potent in expanding CD8+ T cells and NK cells while avoiding expansion of Tregs in a dose dependent manner. ANV419 has a favorable safety profile in non-human primates that separates the targeted anti-tumor immune stimulatory properties from the dose limiting toxicities of recombinant IL-2. This data warrants further translational development of ANV419 as an immune therapeutic in oncology.

SITC Annual Meeting November 2020

Poster #571